SO-C101 induces NK cell cytotoxicity and potentiates antibody-dependent cell cytotoxicity and anti-tumor activity

Zuzana Antosova[1], Nada Podzimkova[1], Katerina Augustynkova[1], Katerina Sajnerova[1], Marketa Jiratova[1], Eva Nedvedova[1], Guy de Martynoff[3], David Bechard[3], Ulrich Moebius[1], Radek Spisek[1,2], Irena Adkins[1,2]

[1]SOTIO a.s, Jankovcova 1518/2, Prague 7, 170 00, Czech Republic; [2]Department of Immunology, 2nd Faculty of Medicine and University Hospital Motol, Charles University, V Uvalu 84, Prague 5, 150 06, Czech Republic; [3]Cytune Pharma, Nantes F-44300, France

Introduction

SO-C101 is a superagonist fusion protein of interleukin (IL)-15 and the IL-15 receptor α (IL-15Rα) sushi+ domain, representing a promising clinical candidate for the treatment of cancer. SO-C101 specifically stimulates natural killer (NK) cells and memory CD8+ T cells with no significant expansion and activation of regulatory T cell compartment. Human NK cell proliferation, the expression of NK cell receptors and ADCC activity of human PBMC after stimulation with SO-C101 *in vitro* in combination with monoclonal antibodies were detected by flow cytometry. The anti-tumor efficacy of SO-C101 in combination with Daratumumab was assessed in a multiple myeloma xenograft mouse model *in vivo*. In this study, we show that SO-C101 induced proliferation and expansion of both major subsets of human NK cells, CD56brightCD16- and CD56dimCD16+. Furthermore, SO-C101 induced expression of the cytotoxic receptors NKp30 and NKG2D whereas no upregulation of the inhibitory receptors CD158a, CD158b and NKG2A was detected. Both NK cell subsets activated by SO-C101 exhibited cytotoxicity towards cancer cells *in vitro*. Using human PBMCs, we show that SO-C101 potentiated killing of tumor cells induced by several clinically approved therapeutic monoclonal antibodies such as Cetuximab, Daratumumab and Obinutuzumab *in vitro*. SO-C101 and Daratumumab monotherapy treatment inhibited tumor growth *in vivo*, however, their combination showed the strongest anti-tumor efficacy. Specifically, sequential administration of Daratumumab, followed by SO-C101 promoted complete tumor regression, compared to partial anti-tumor responses induced by the respective monotherapies. SO-C101 augments the anti-tumor activity of therapeutic antibodies by increasing NK cells mediated antibody-dependent cell cytotoxicity. These results support the evaluation of SO-C101 in combination with monoclonal therapeutic antibodies in clinical studies.

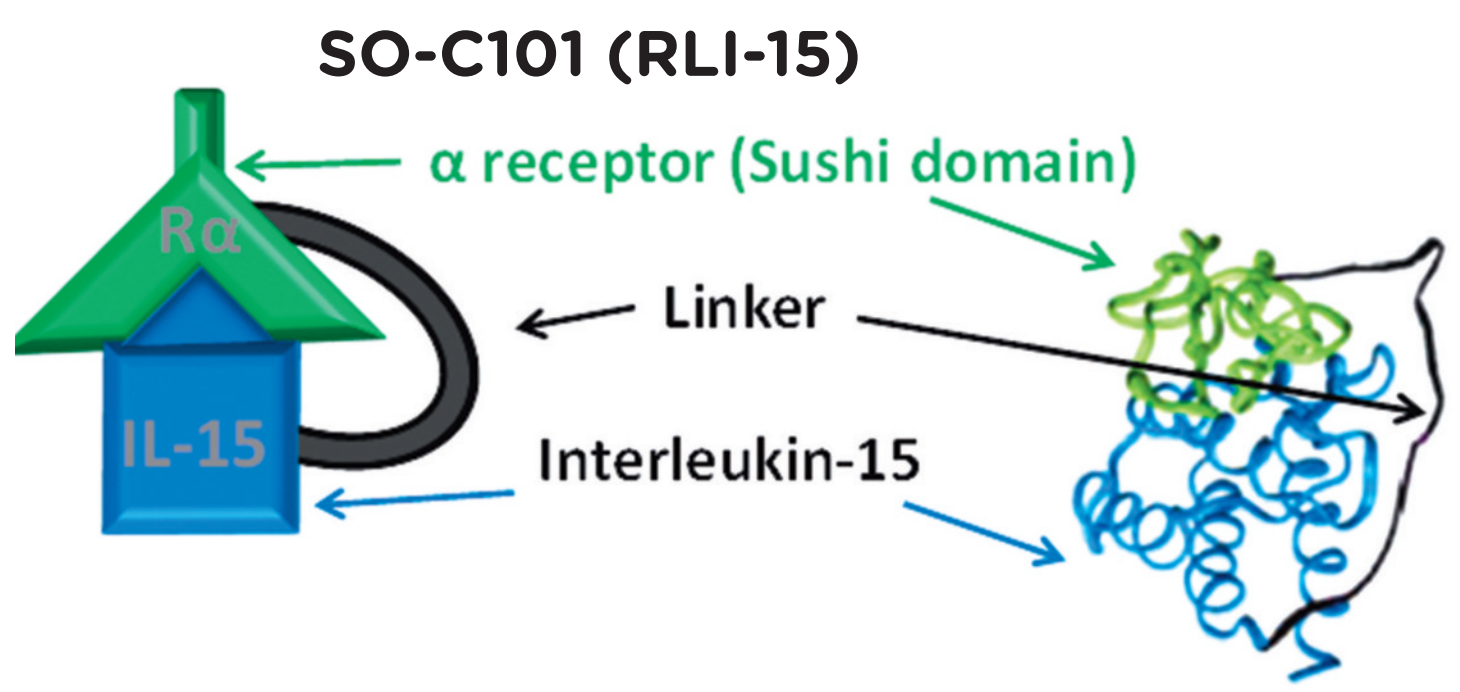

SO-C101 (RLI-15)
α receptor (Sushi domain)
Rα
Linker
IL-15
Interleukin-15

- SO-C101 (211 amino acids) is a fusion protein that consists of the N-terminal sushi+ domain of the human IL-15Rα (77 amino acids) covalently coupled via a non-immunogenic linker of Glycine-Serine residues (20 amino acids) to the mature IL-15 sequence (114 amino acids) (Mortier et al., 2006).
- SO-C101 acts as a selective and potent agonist of the IL-15 pathway through IL-15Rβγ thereby inducing proliferation and activation of memory CD8+ T cells, NK cells, γδ T cells and NKT cells *in vitro* and *in vivo* and exerts significantly increased anti-tumor efficacy in various mouse cancer models over IL-15 (Bessard et al., 2009; Desbois et al., 2016)

Figure 1

SO-C101 expands and activates human NK cells *in vitro*

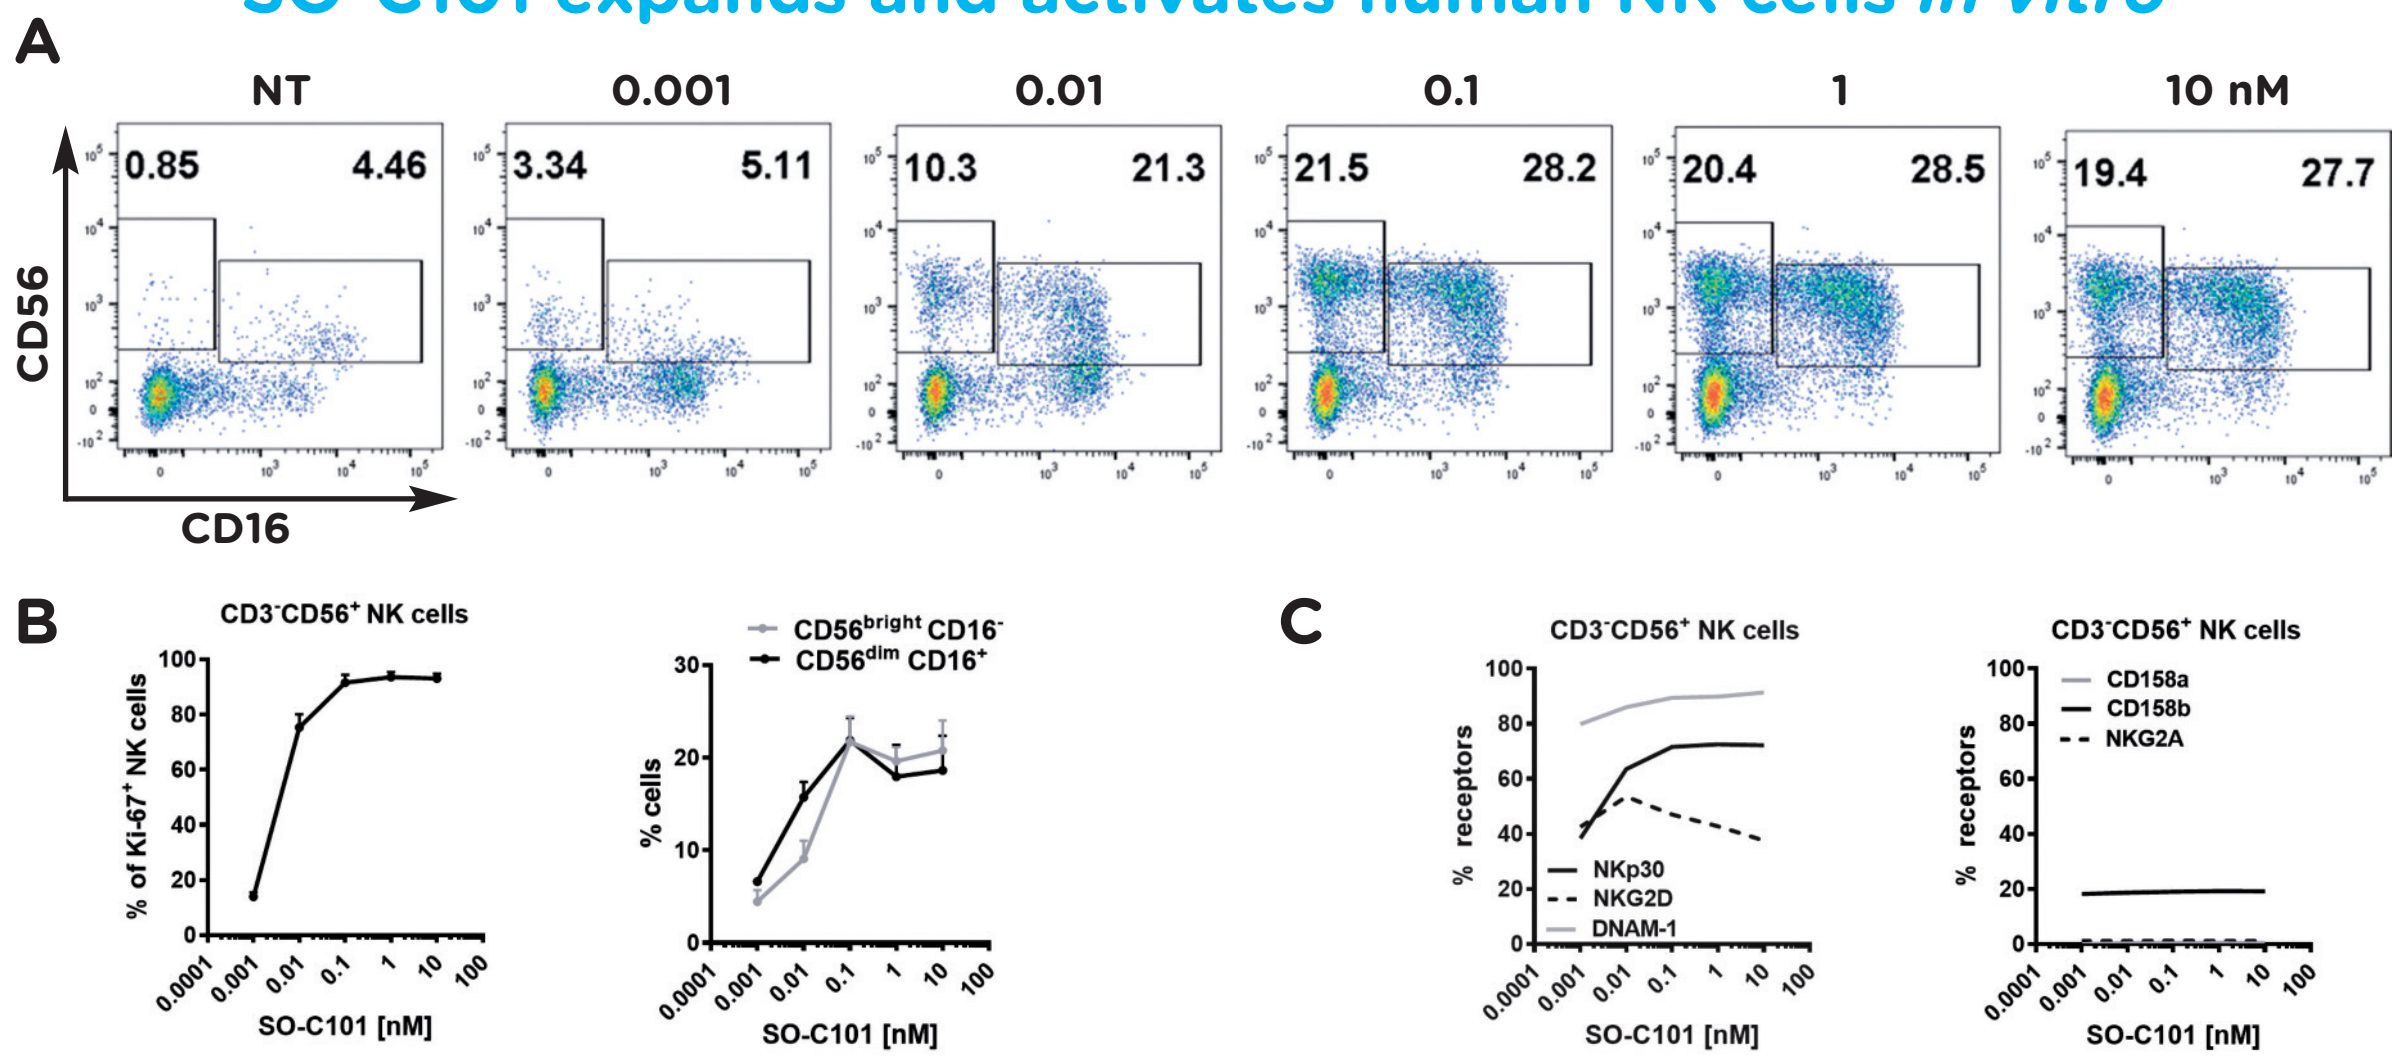

A
NT
0.001
0.01
0.1
1
10 nM
0.85
4.46
3.34
5.11
10.3
21.3
21.5
28.2
20.4
28.5
19.4
27.7
CD56
CD16
B
CD3-CD56+ NK cells
% of Ki-67+ NK cells
CD56bright CD16-
CD56dim CD16+
% cells
SO-C101 [nM]
C
CD3-CD56+ NK cells
% receptors
NKp30
NKG2D
DNAM-1
CD158a
CD158b
NKG2A

Figure 1. Human PBMCs were incubated with the indicated concentrations of SO-C101 for 7 days (NT – non-treated) and NK cells (CD3-CD56+) were analyzed by flow cytometry. **(A, B)** SO-C101 stimulates and expands human NK cells and their subpopulations CD56dimCD16+ and CD56brightCD16- in a concentration-dependent manner *in vitro*. NK cell proliferation was detected as the percentage of Ki67+ cells **(C)** SO-C101 induces the expression of activation receptors (DNAM-1, NKp30 and NKG2D) on NK cells without inducing the expression of the inhibitory receptors (CD158a, CD158b and NKG2A) after 7 days of stimulation *in vitro*. Data represent mean + SEM from 4 healthy donors.

Figure 2

SO-C101 stimulates cytotoxic activity of human NK cells *in vitro*

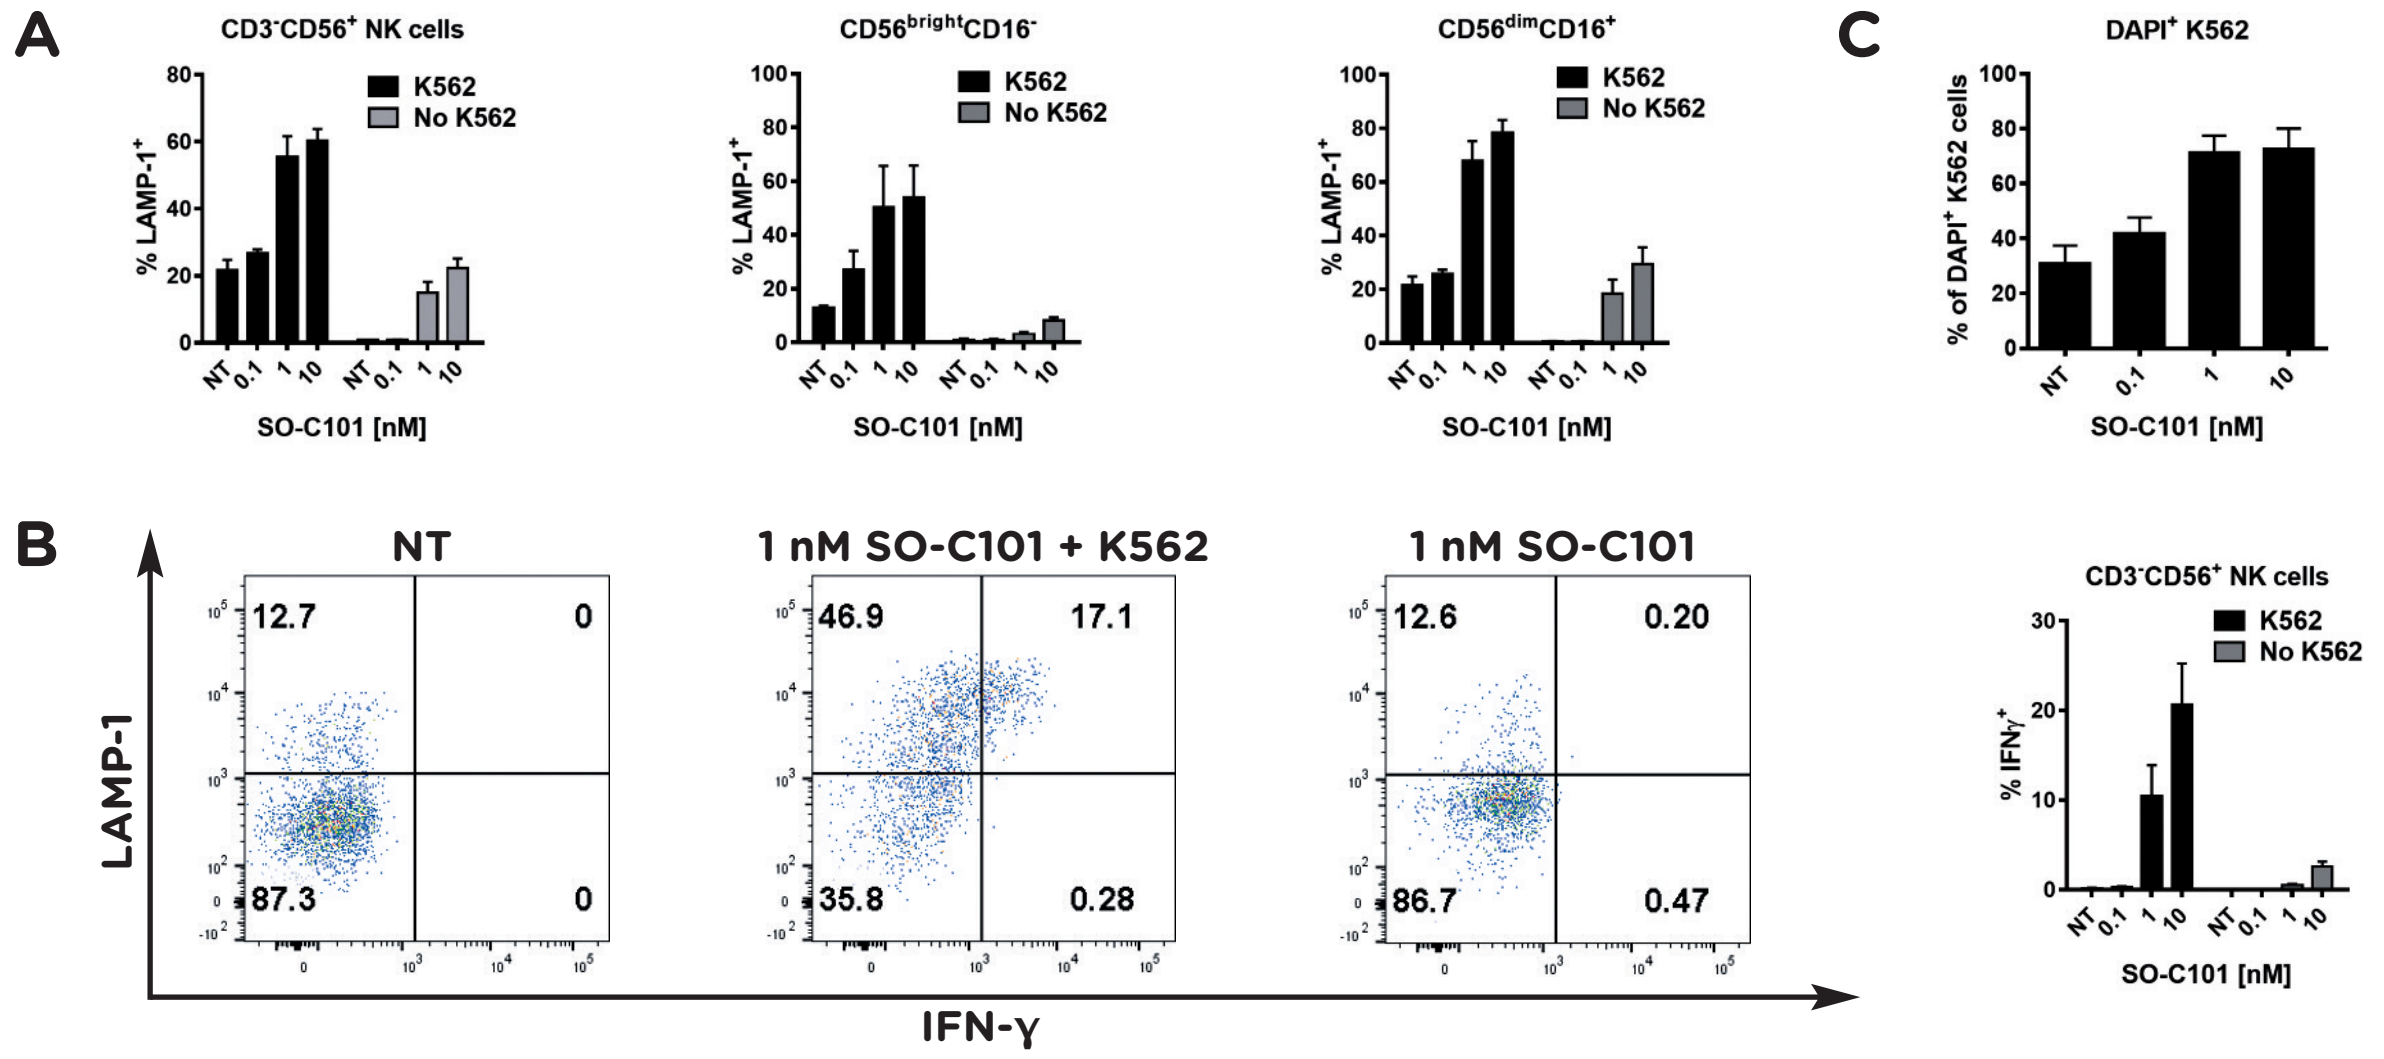

A
CD3-CD56+ NK cells
CD56brightCD16-
CD56dimCD16+
K562
No K562
% LAMP-1+
SO-C101 [nM]
C
DAPI+ K562
% of DAPI+ K562 cells
B
NT
1 nM SO-C101 + K562
1 nM SO-C101
12.7
0
87.3
0
46.9
17.1
35.8
0.28
12.6
0.20
86.7
0.47
LAMP-1
IFN-γ
% IFNγ+

Figure 2. The cytotoxic activity of human NK cells induced by SO-C101 was analyzed by flow cytometry using LAMP-1 staining as a marker of cytotoxic degranulation, intracellular IFN-γ staining and detection of dead tumor cells (DAPI+). **(A)** NK cells and their both subsets CD56dimCD16+ and CD56brightCD16- exhibit cytotoxic activity towards K562 tumor cells. **(B)** Representative dotplots of LAMP-1 and IFN-γ staining in NK cells are shown (NT – non-treated; 1 nM SO-C101 + K562 – human PBMC treated with SO-C101 and co-cultivated with K562 target cells; 1 nM SO-C101 – human PBMC treated with SO-C101 without K562 co-cultivation). **(C)** The percentage of dead DAPI+ K562 tumor cells was determined by flow cytometry after 6 h treatment. Data represent mean ± SEM from 4 healthy donors.

Figure 3

SO-C101 enhances the activity of therapeutic antibodies in human PBMC killing assay *in vitro*

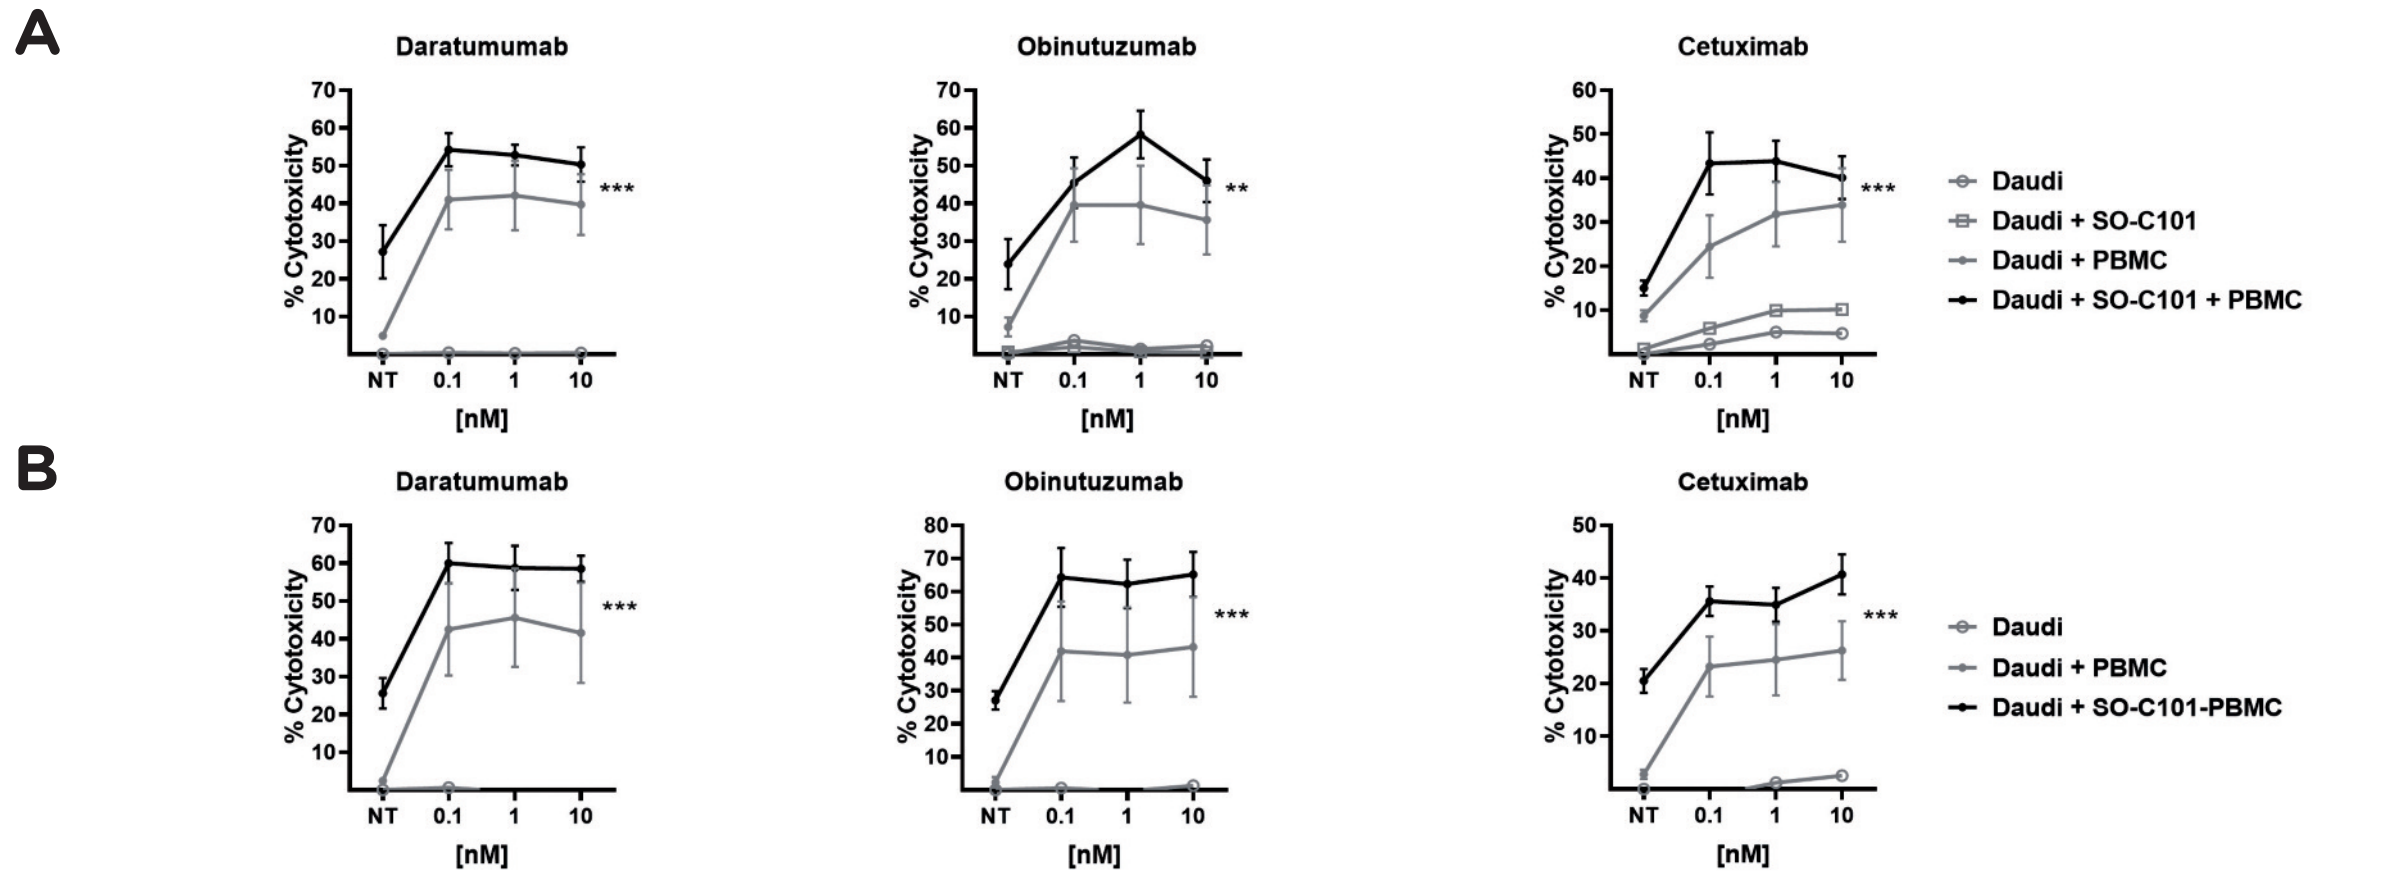

A
Daratumumab
Obinutuzumab
Cetuximab
% Cytotoxicity
[nM]
Daudi
Daudi + SO-C101
Daudi + PBMC
Daudi + SO-C101 + PBMC
B
Daudi
Daudi + PBMC
Daudi + SO-C101-PBMC

Figure 3. SO-C101 potentiates the activity of Daratumumab, Obinutuzumab and Cetuximab to kill tumor cells in a concomitant (A) and sequential (B) treatment setting in human PBMC killing assay *in vitro*. Percentage of cytotoxicity was determined by a lactate dehydrogenase release assay (Cytotoxicity Detection Kit). **(A) Concomitant treatment.** Human PBMCs were added to the target tumor cells together with 1 nM SO-C101 and the therapeutic antibodies at the indicated concentrations and incubated for 20 h. E:T ratio was 25:1 for Daudi (Daratumumab, Obinutuzumab) and 150:1 for SCC25 (Cetuximab) cells, respectively. **(B) Sequential treatment.** Human PBMCs were first stimulated with 0.1 nM SO-C101 for 2 days (SO-C101-PBMC) and then added to the target tumor cells together with therapeutic antibodies at the indicated concentrations and incubated for 24 h. E:T ratio was 15:1 for Daudi and 100:1 for SCC25, respectively. Data represent mean ± SEM from 3 healthy donors. **P < 0.0021, ***P < 0.0002, Wilcoxon signed rank test. (NT – non-treated).

Figure 4

SO-C101 stimulated NK cells potentiate antibody-dependent cell cytotoxicity (ADCC) *in vitro*

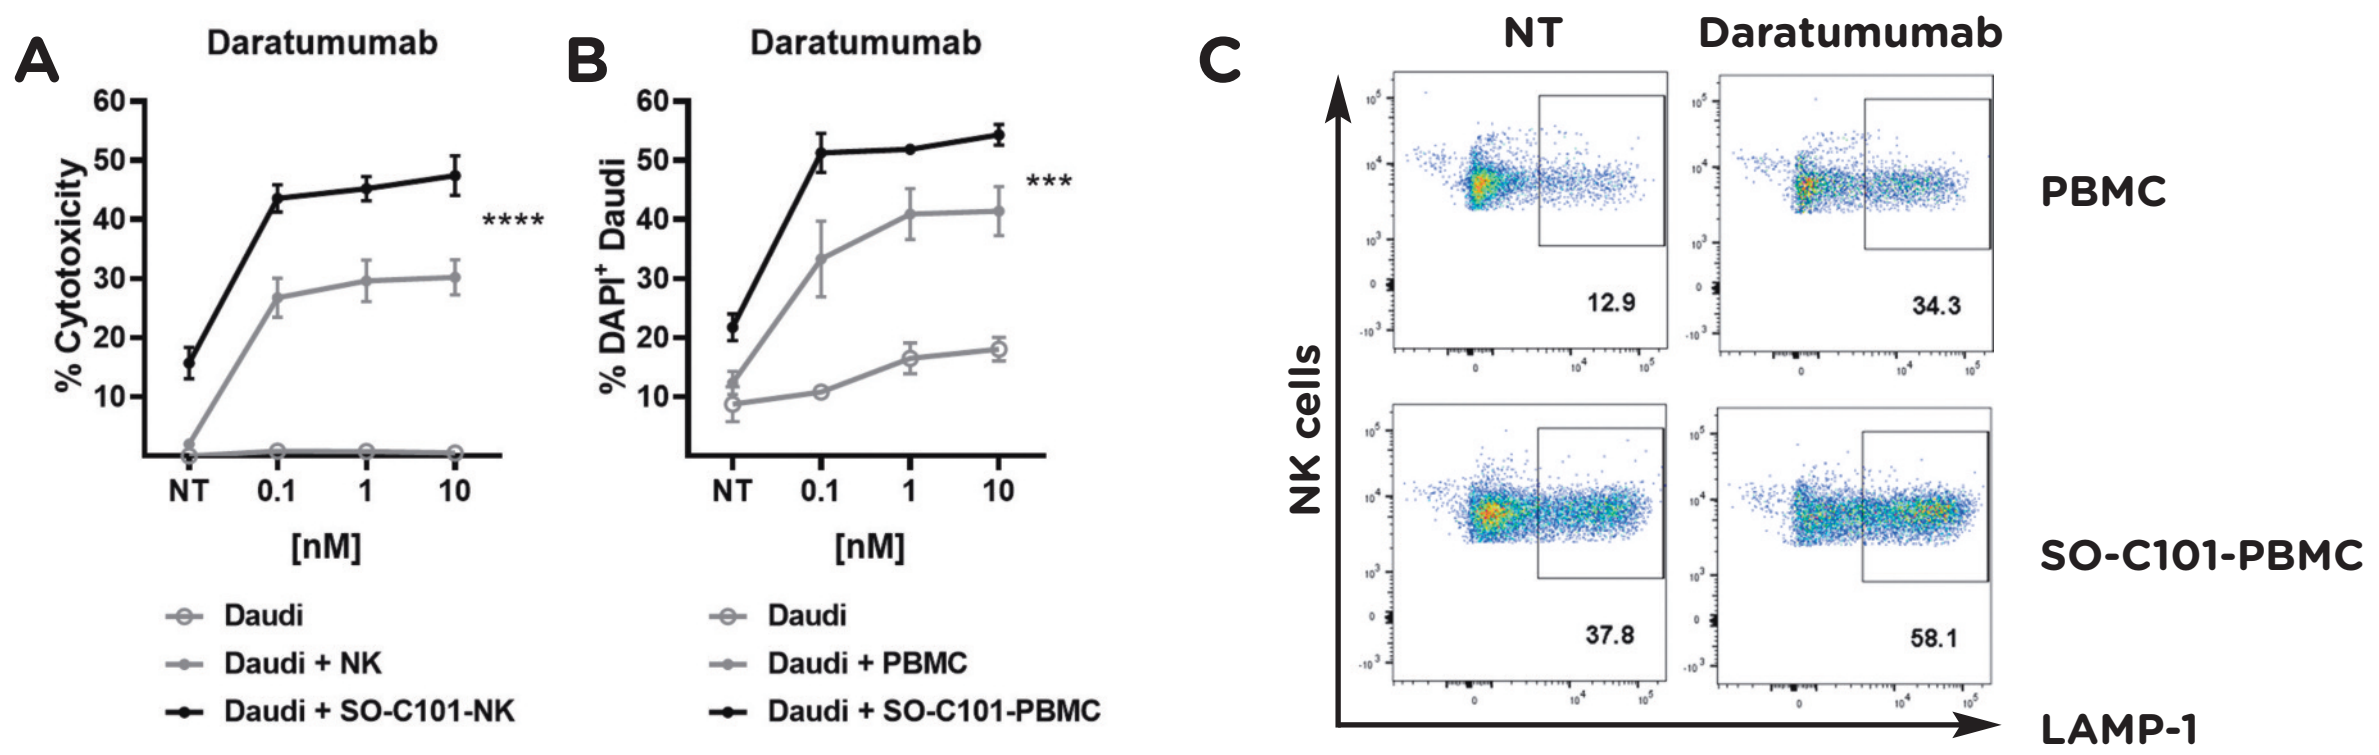

A
Daratumumab
% Cytotoxicity
[nM]
Daudi
Daudi + NK
Daudi + SO-C101-NK
B
Daratumumab
% DAPI+ Daudi
Daudi
Daudi + PBMC
Daudi + SO-C101-PBMC
C
NT
Daratumumab
PBMC
SO-C101-PBMC
NK cells
LAMP-1
12.9
34.3
37.8
58.1

Figure 4. SO-C101 stimulation of human NK cells (A) and PBMCs (B, C) potentiates antibody-dependent cell cytotoxicity (ADCC) of Daratumumab. **(A)** Isolated human NK cells were treated with SO-C101 (0.1 nM, 3 days) and then incubated with Daratumumab and Daudi tumor cells (E: T 10:1) for 4 h. Cytotoxicity was determined by a lactate dehydrogenase release assay **(B)** Human PBMCs were treated with SO-C101 (0.1 nM, 2 days) and then incubated with Daratumumab and Daudi cells (E:T 1:1) for 4 h. Tumor cell death (DAPI+) was detected by flow cytometry. **(C)** Furthermore the cytotoxic activity of human NK cells was determined by LAMP-1 staining as a marker of cytotoxic degranulation. Representative dot plots are shown, where Daratumumab was used at 0.1 nM. Data represent mean ± SEM from 6 (A) or 3 (B, C) healthy donors. ****P < 0.0001, ***P < 0.0002, Wilcoxon signed rank test.

Figure 5

SO-C101 potentiates therapeutic activity of Daratumumab *in vivo*

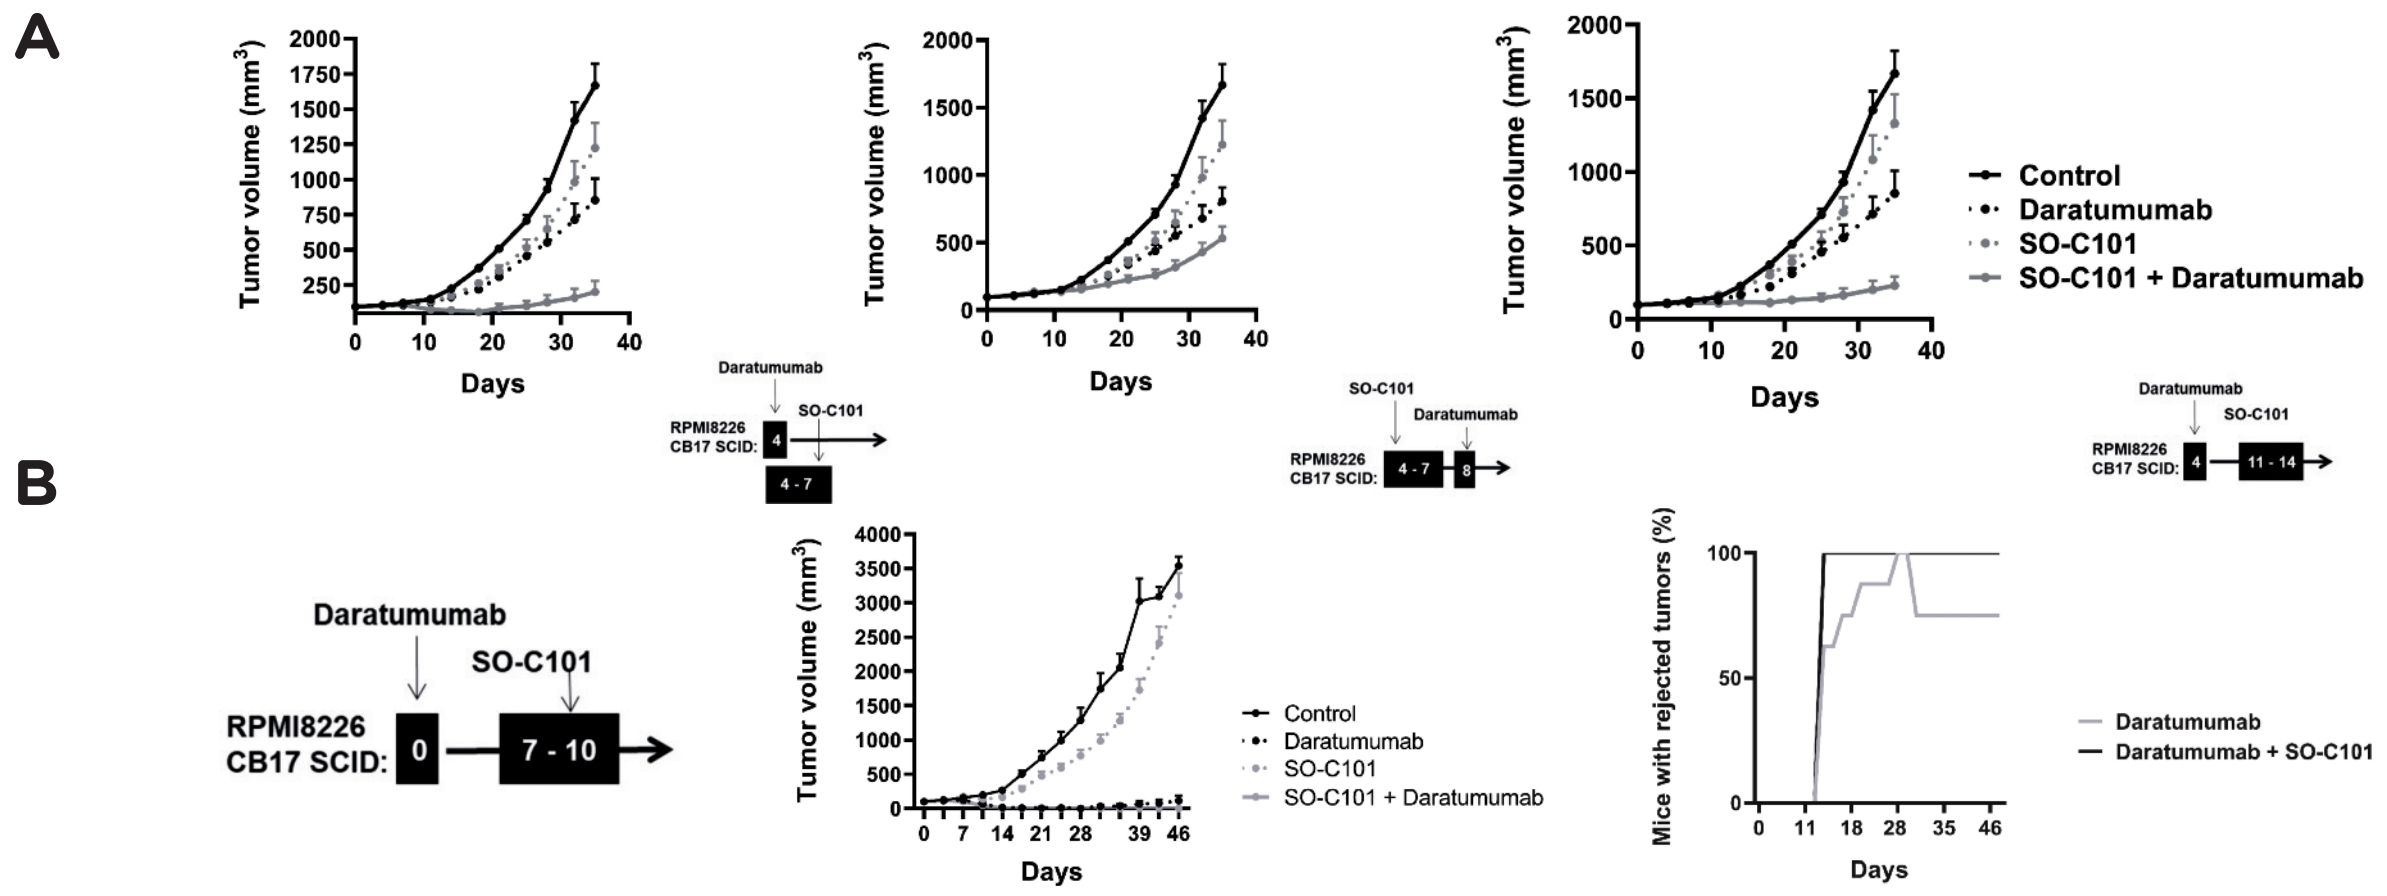

A
Tumor volume (mm3)
Days
Control
Daratumumab
SO-C101
SO-C101 + Daratumumab
RPMI8226 CB17 SCID:
B
Daratumumab
SO-C101
RPMI8226 CB17 SCID:
0
7 - 10
Mice with rejected tumors (%)
Daratumumab + SO-C101

Figure 5. (A) SO-C101 and Daratumumab treatment decreases the tumor growth in human multiple myeloma Xenograft model (RPMI8226) in CB17 SCID mice. Mice were treated s.c with SO-C101 at 1 mg/kg and i.p. with Daratumumab at 20 mg/kg according to the schemes. **(B)** Co-administration of SO-C101 (1 mg/kg, day 7-10) and Daratumumab (20 mg/kg, day 0) demonstrated the most potent anti-tumor activity with tumor growth inhibition (TGI) value of 100 %. Data are presented as mean + SEM from 8 mice.

Conclusions

- **SO-C101 expands and activates both subsets of human NK cells *in vitro* which is accompanied by the expression of the activation receptors**
- **SO-C101 stimulates the cytotoxic activity of human NK cells towards tumor cells *in vitro***
- **SO-C101 enhances the activity of Daratumumab, Obinutuzumab and Cetuximab to kill tumor cells in human PBMC killing assay *in vitro*.**
- **SO-C101 potentiates antibody dependent cell cytotoxicity (ADCC) of Daratumumab *in vitro*.**
- **SO-C101 synergizes with Daratumumab in anti-tumor efficacy *in vivo*.**

For more information please contact Irena Adkins, adkins@sotio.com